Red-Light Photodynamic Therapy with 10% ALA Gel for the Treatment of Keratoacanthoma: A Prospective Pilot Study

Jeffrey Cizenski, MD • Karah Blair, BS *Empire Dermatology | Syracuse, NY*

KEY FINDINGS

100% CLINICAL CLEARANCE
Complete clinical resolution in all subjects at 3 mos

0 RECURRENCES
No clinical recurrence observed at 3 mos

WELL TOLERATED
Low pain scores during illumination

Before and After lesion of a representative subject

BEFORE TREATMENT | **3-MONTH FOLLOW-UP**

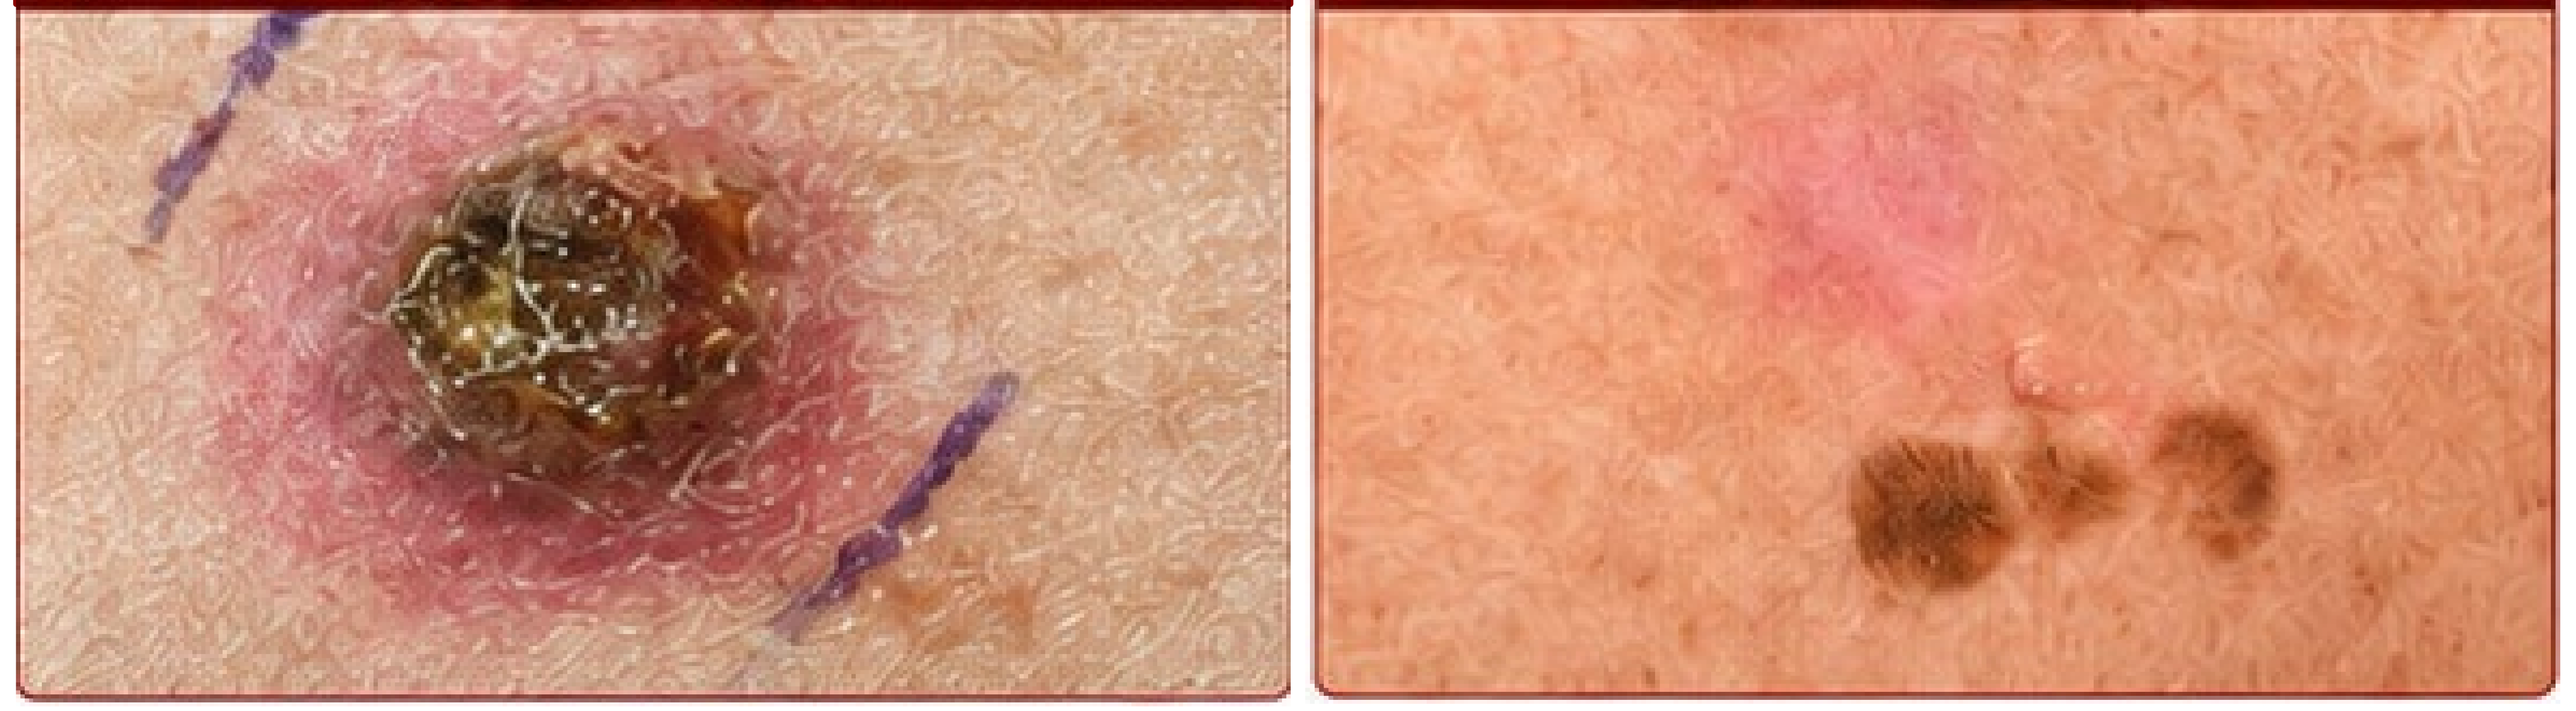

OBJECTIVE

Primary Objective: to determine the efficacy of red-light photodynamic therapy (PDT) with 10% ALA gel for treatment of keratoacanthoma (KA) on the trunk and extremities.
Secondary objective: to evaluate tolerability during illumination using patient-reported VAS scores.

STUDY AT A GLANCE

n = 5 subjects → PDT #1 → 2 weeks apart → PDT #2 → 3-month f/u

INTRODUCTION

KA is a rapidly growing, well-differentiated squamous neoplasm generally considered a variant of squamous cell carcinoma (SCC). Although some lesions may undergo spontaneous regression, their unpredictable clinical behavior and histopathologic overlap with invasive SCC typically warrant definitive treatment. Surgical excision remains the standard of care; however, surgery may not be ideal for all patients because of lesion size, location, cosmetic considerations, medical comorbidities, or patient preference.
Red-light PDT with 10% ALA gel is FDA approved for the treatment of actinic keratoses (AKs) of the face and scalp. PDT is a minimally invasive, tissue-selective treatment that combines topical ALA with red-light illumination to generate reactive oxygen species, resulting in selective destruction of dysplastic and neoplastic cells while preserving surrounding healthy tissue. Numerous studies have demonstrated its efficacy in superficial basal cell carcinoma (BCC) and squamous cell carcinoma in situ (SCCis).
Although several published case reports have described successful treatment of eruptive keratoacanthoma with PDT, no prospective clinical studies have systematically evaluated the safety, tolerability, and efficacy of red-light PDT with 10% ALA gel for KA.
This prospective pilot study was therefore conducted to evaluate the clinical response, safety, and cosmetic outcomes of red-light photodynamic therapy using 10% ALA gel in patients with biopsy-confirmed KA.

LESION SIZE (cm) AT EACH VISIT

↗ Progressive reduction in lesion size observed across treatments, with complete clearance maintained at 3-month follow-up

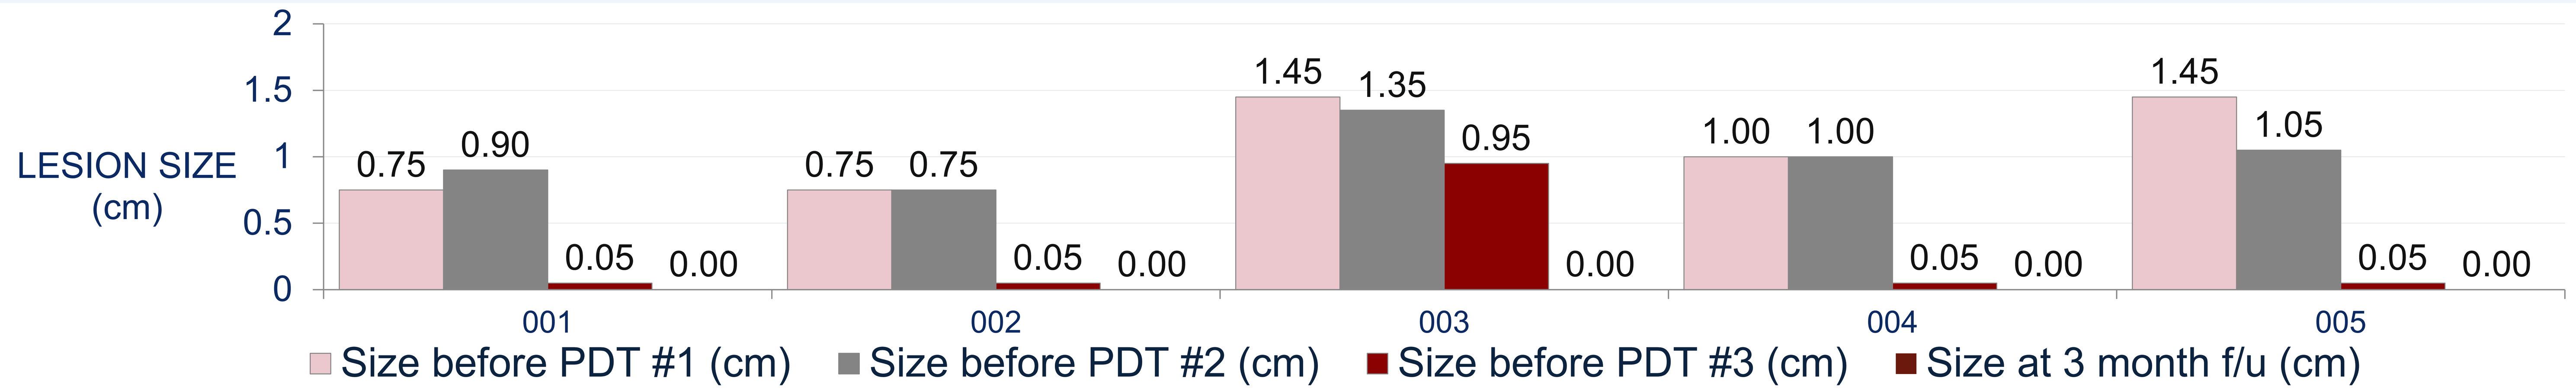
LESION SIZE (cm)
2
1.5
1
0.5
0
0.75 0.90 0.05 0.00
0.75 0.75 0.05 0.00
1.45 1.35 0.95 0.00
1.00 1.00 0.05 0.00
1.45 1.05 0.05 0.00
001
002
003
004
005
Size before PDT #1 (cm)
Size before PDT #2 (cm)
Size before PDT #3 (cm)
Size at 3 month f/u (cm)

✓ All subjects achieved complete clinical clearance by 3 months and maintained clearance

METHODS

Study population: Five subjects with 0.5–1.5 cm KA or well-differentiated SCC on the trunk or extremities
Two PDT performed 2 weeks apart; one subject received a third treatment as clinically indicated

PDT PROTOCOL

1. LESION DEBRIDEMENT →

2. ALA APPLICATION
10% ALA gel applied to lesion →

3. INCUBATION
2.5 hr incubation under occlusion →

4. ILLUMINATION
Red light, 630 nm, 37 J/cm2

MEAN VAS PER PDT TREATMENT

Pain scores were generally low during illumination, with mean VAS ≤ 3.3 for the first two treatments

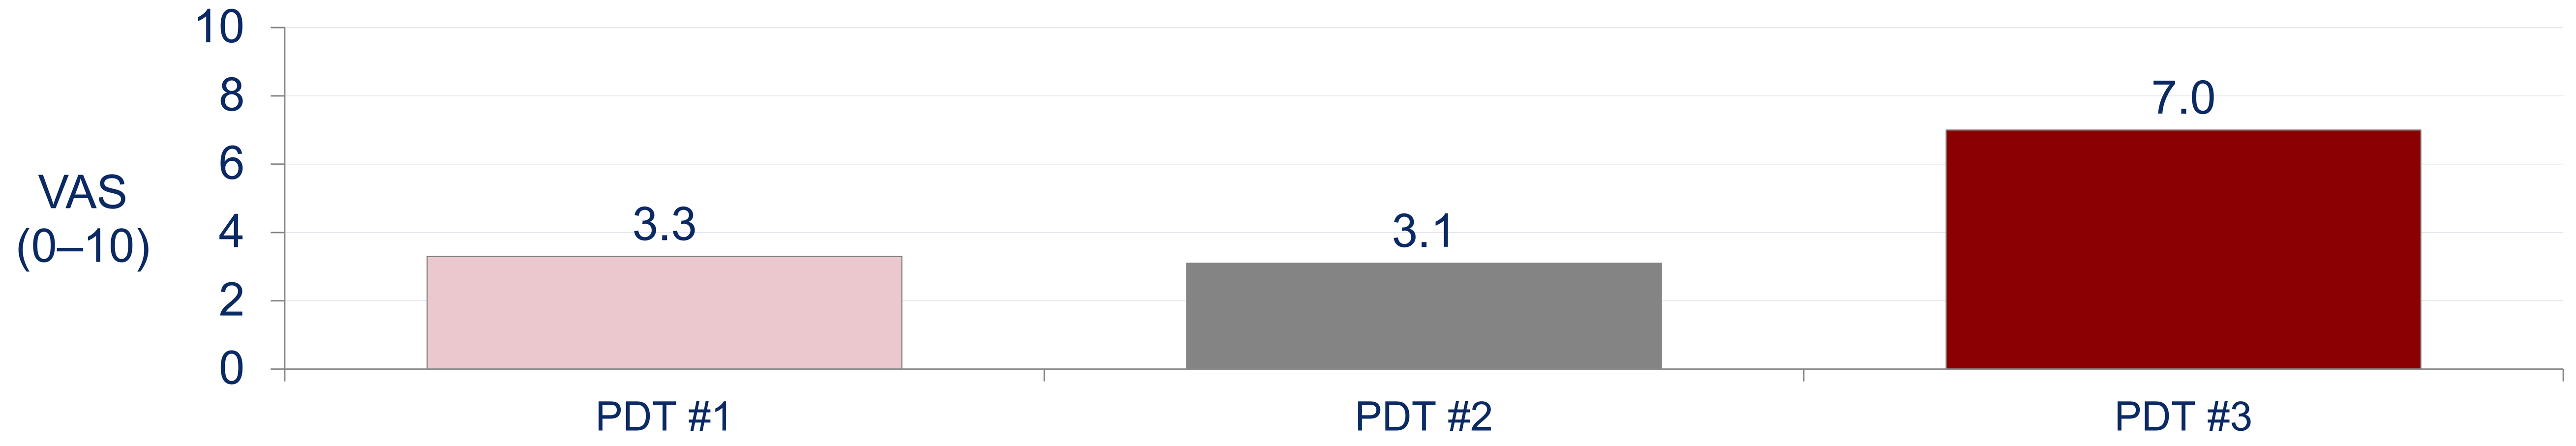
VAS (0–10)
10
8
6
4
2
0
3.3
3.1
7.0
PDT #1
PDT #2
PDT #3

Scale: 0 = No pain • 10 = Worst pain imaginable *VAS = Visual Analog Scale*

RESULTS

- All subjects showed complete clinical resolution at 3-month follow-up
- All subjects had a lesion score of 0 at 3-month follow-up
- No recurrences were documented during follow-up
- Mean pain scores were:
 3.3 during PDT #1
 3.0 during PDT #2
 7.0 during PDT #3
- Lesion size decreased to 0 cm at 3-month follow-up for all subjects

CONCLUSION

- Red-light PDT with 10% ALA gel demonstrated complete clinical resolution of KA in this pilot series
- Treatment was generally well tolerated
- No clinical recurrence was observed at 3-month follow-up
- Larger prospective studies with longer follow-up are warranted

REFERENCES

1. Ameluz [prescribing information]. Biofrontera Inc; 2024.
2. Schlesinger T, et al. J Am Acad Dermatol. 2025;S0190-9622(25)02656-8.
3. Morton CA, et al. Br J Dermatol. 2018;179(2):308–319.
4. Cervantes JA, Zeitouni NC. Photodiagnosis Photodyn Ther. 2021;35:102358.
5. Shim PJ, Zeitouni NC. Photodiagnosis Photodyn Ther. 2023;41:103211.
6. Liang X, Lin S, Yan J. Photodiagnosis Photodyn Ther. 2020;30:101798.
7. Radakovic-Fijan S, et al. Br J Dermatol. 1999;141(S):936–938.
8. Ly T, et al. Photodiagnosis Photodyn Ther. 2018;23:151–153.
9. Alloo A, et al. Arch Dermatol. 2012;148(3):363–366.

This study was investigator-initiated and funded by Biofrontera Inc. The authors have no financial relationships to disclose.